Rapid Phenotypic Detection of Gram-negative Bacilli Resistant to Oximinocephalosporins and Carbapenems in Positive Blood Cultures Using a Novel Protocol

Diego Josa M.[1], Gisell Bustos M.[1,4] Soad Yusef [2,4], Stephanie Crevoisier [2,4], Edwin Silva[1,3], Natalia López[1], Rafael Leal[1], Isabel Torres M.[1] , Juan Osorio[1,3], Gerson Arias[1,3] and Luis Felipe Reyes[4]
1, Research Group Cardiovascular Medicine and Specialties of High Complexity, Fundación Clínica Shaio, Colombia; 2, Department of Critical Medicine, Fundación Clínica Shaio, Colombia; 3, Infectious Diseases Department, Fundación Clínica Shaio, Colombia; 4, Universidad de La Sabana, Colombia.

BACKGROUND

- The initiation of effective antibiotic therapy early improves the survival of patients with Bloodstream infections (BSI) (1).
- Up to 40% of patients with BSI due to multi-drug resistant pathogens (MDRP) die (2).
- The microbiological characterization of MDRP by current conventional techniques can take up to 96 hours.
- We designed a modified protocol to detect MDR Gram-negative bacilli from positive blood cultures using the HB&L® system.

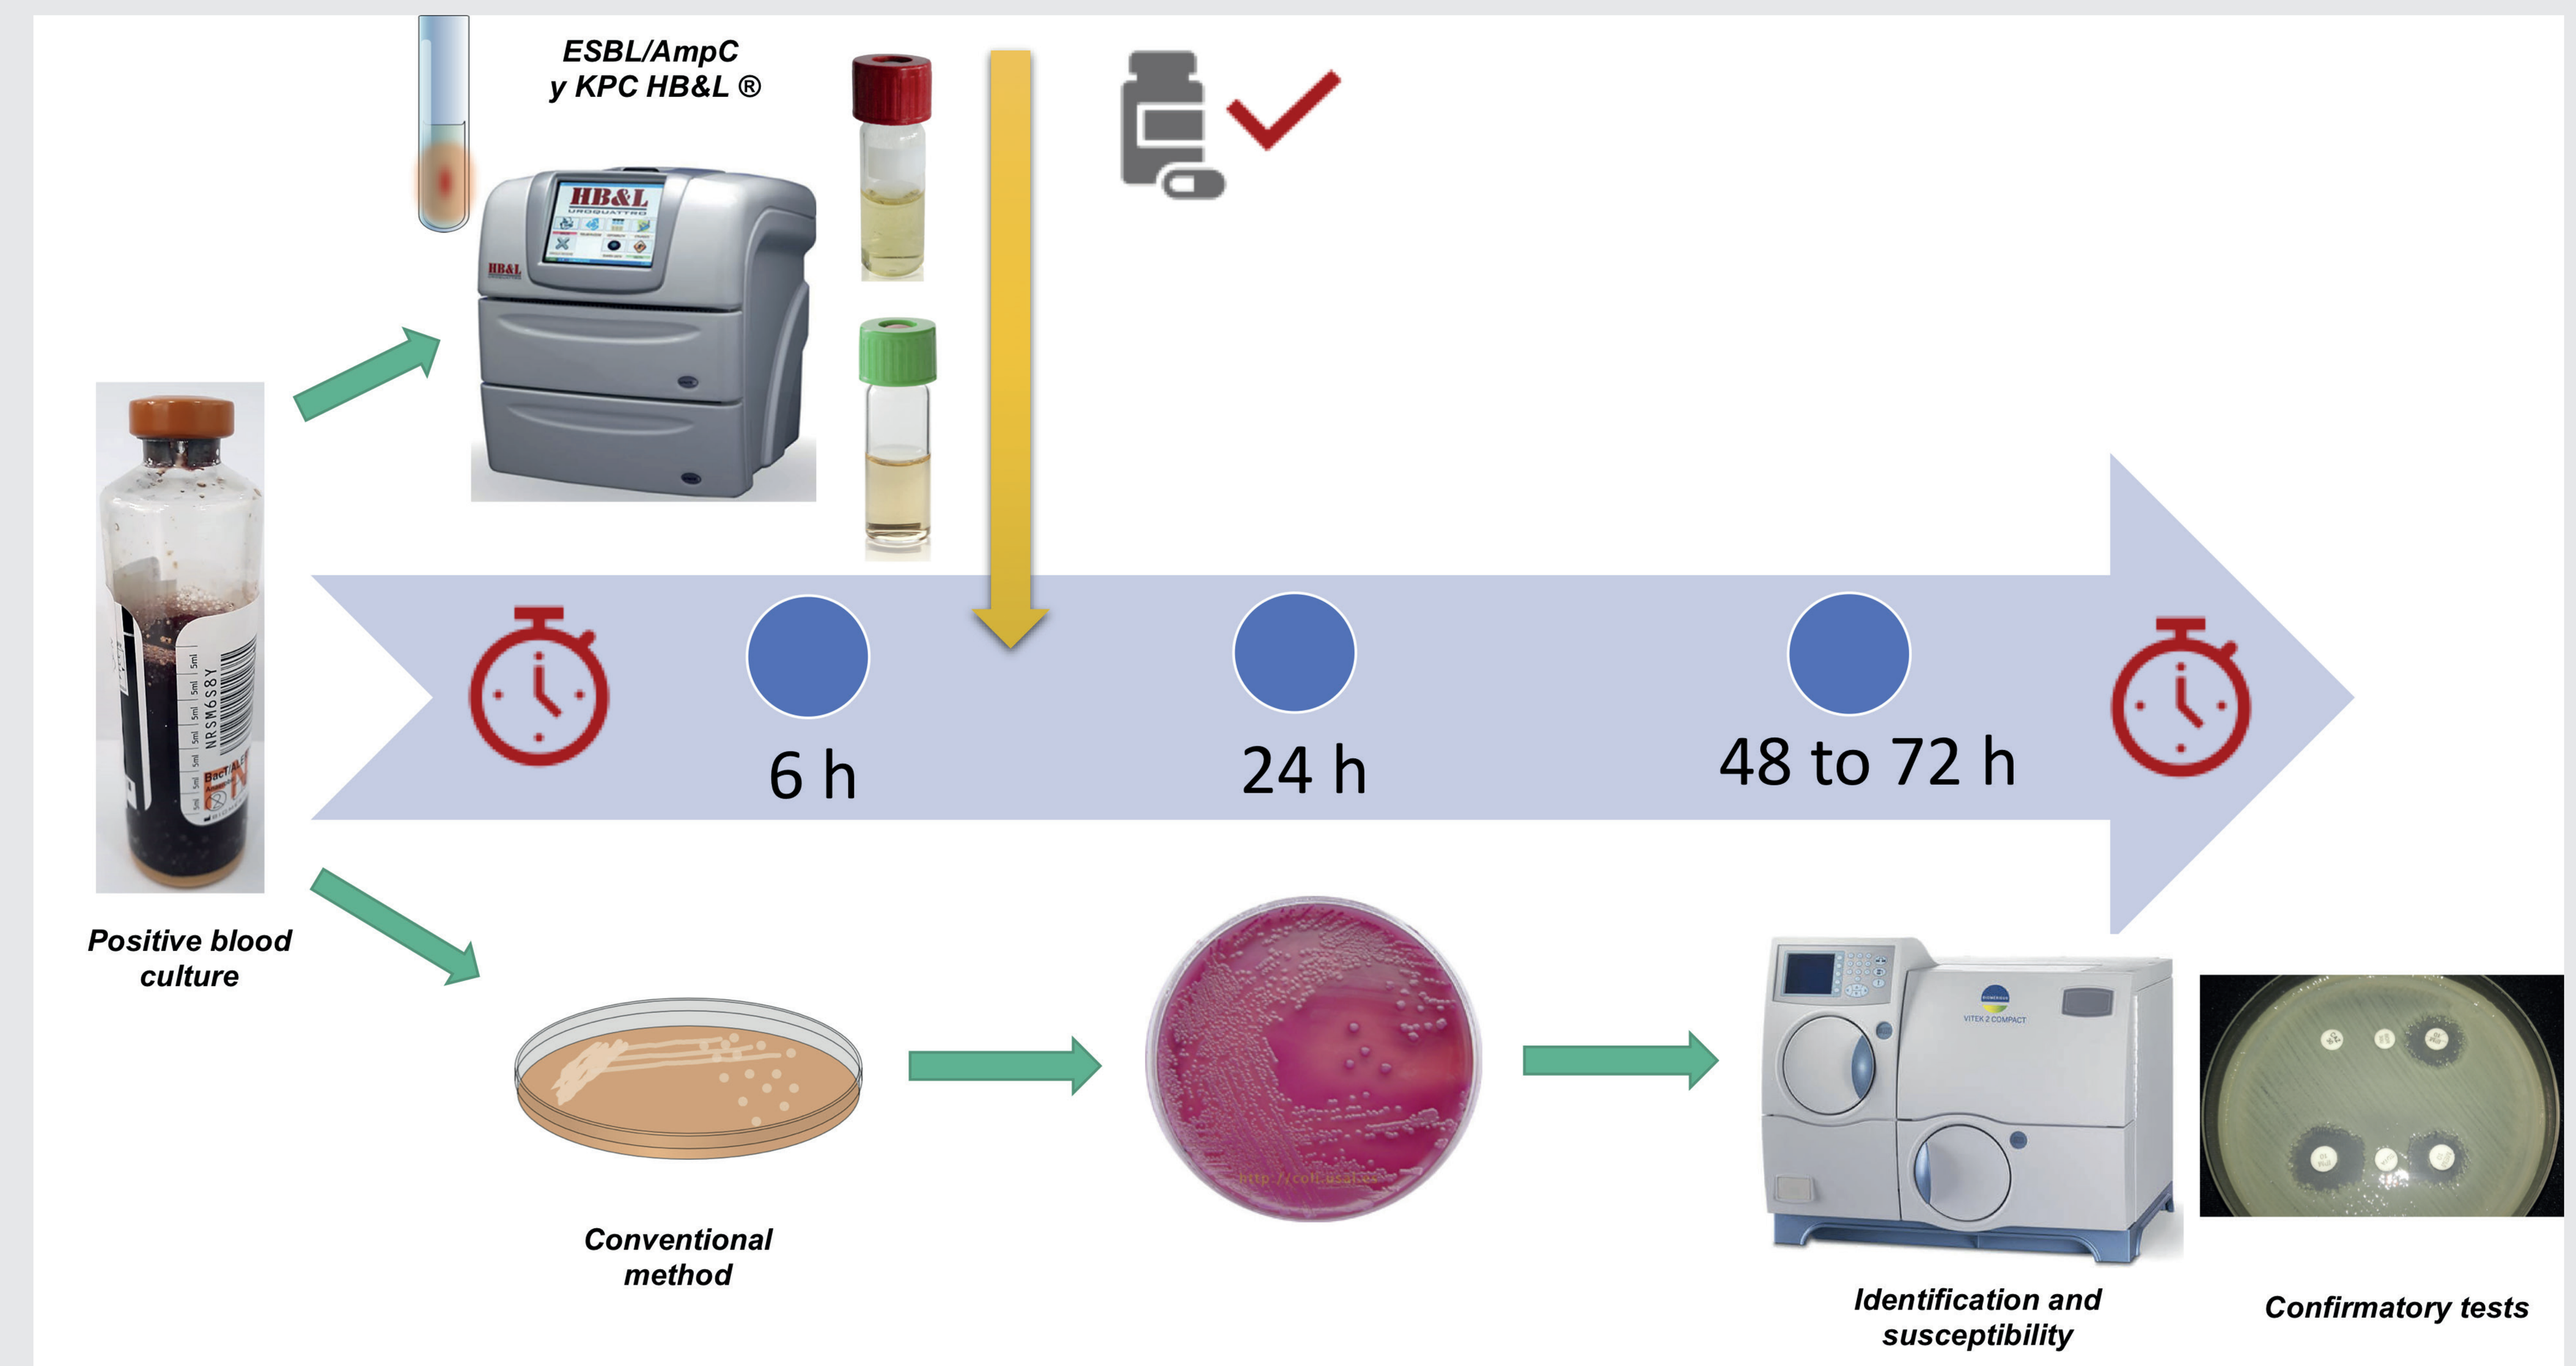
ESBL/AmpC
y KPC HB&L ®
6 h
24 h
48 to 72 h
Positive blood culture
Conventional method
Identification and susceptibility
Confirmatory tests

METHODS

- Observational study, prospective, diagnostic test
- ESBL/AmpC HB&L® test and Carbapenemase kit HB&L® test were evaluated in blood cultures
- Operational characteristics, concordance and reduction of identification time were calculated

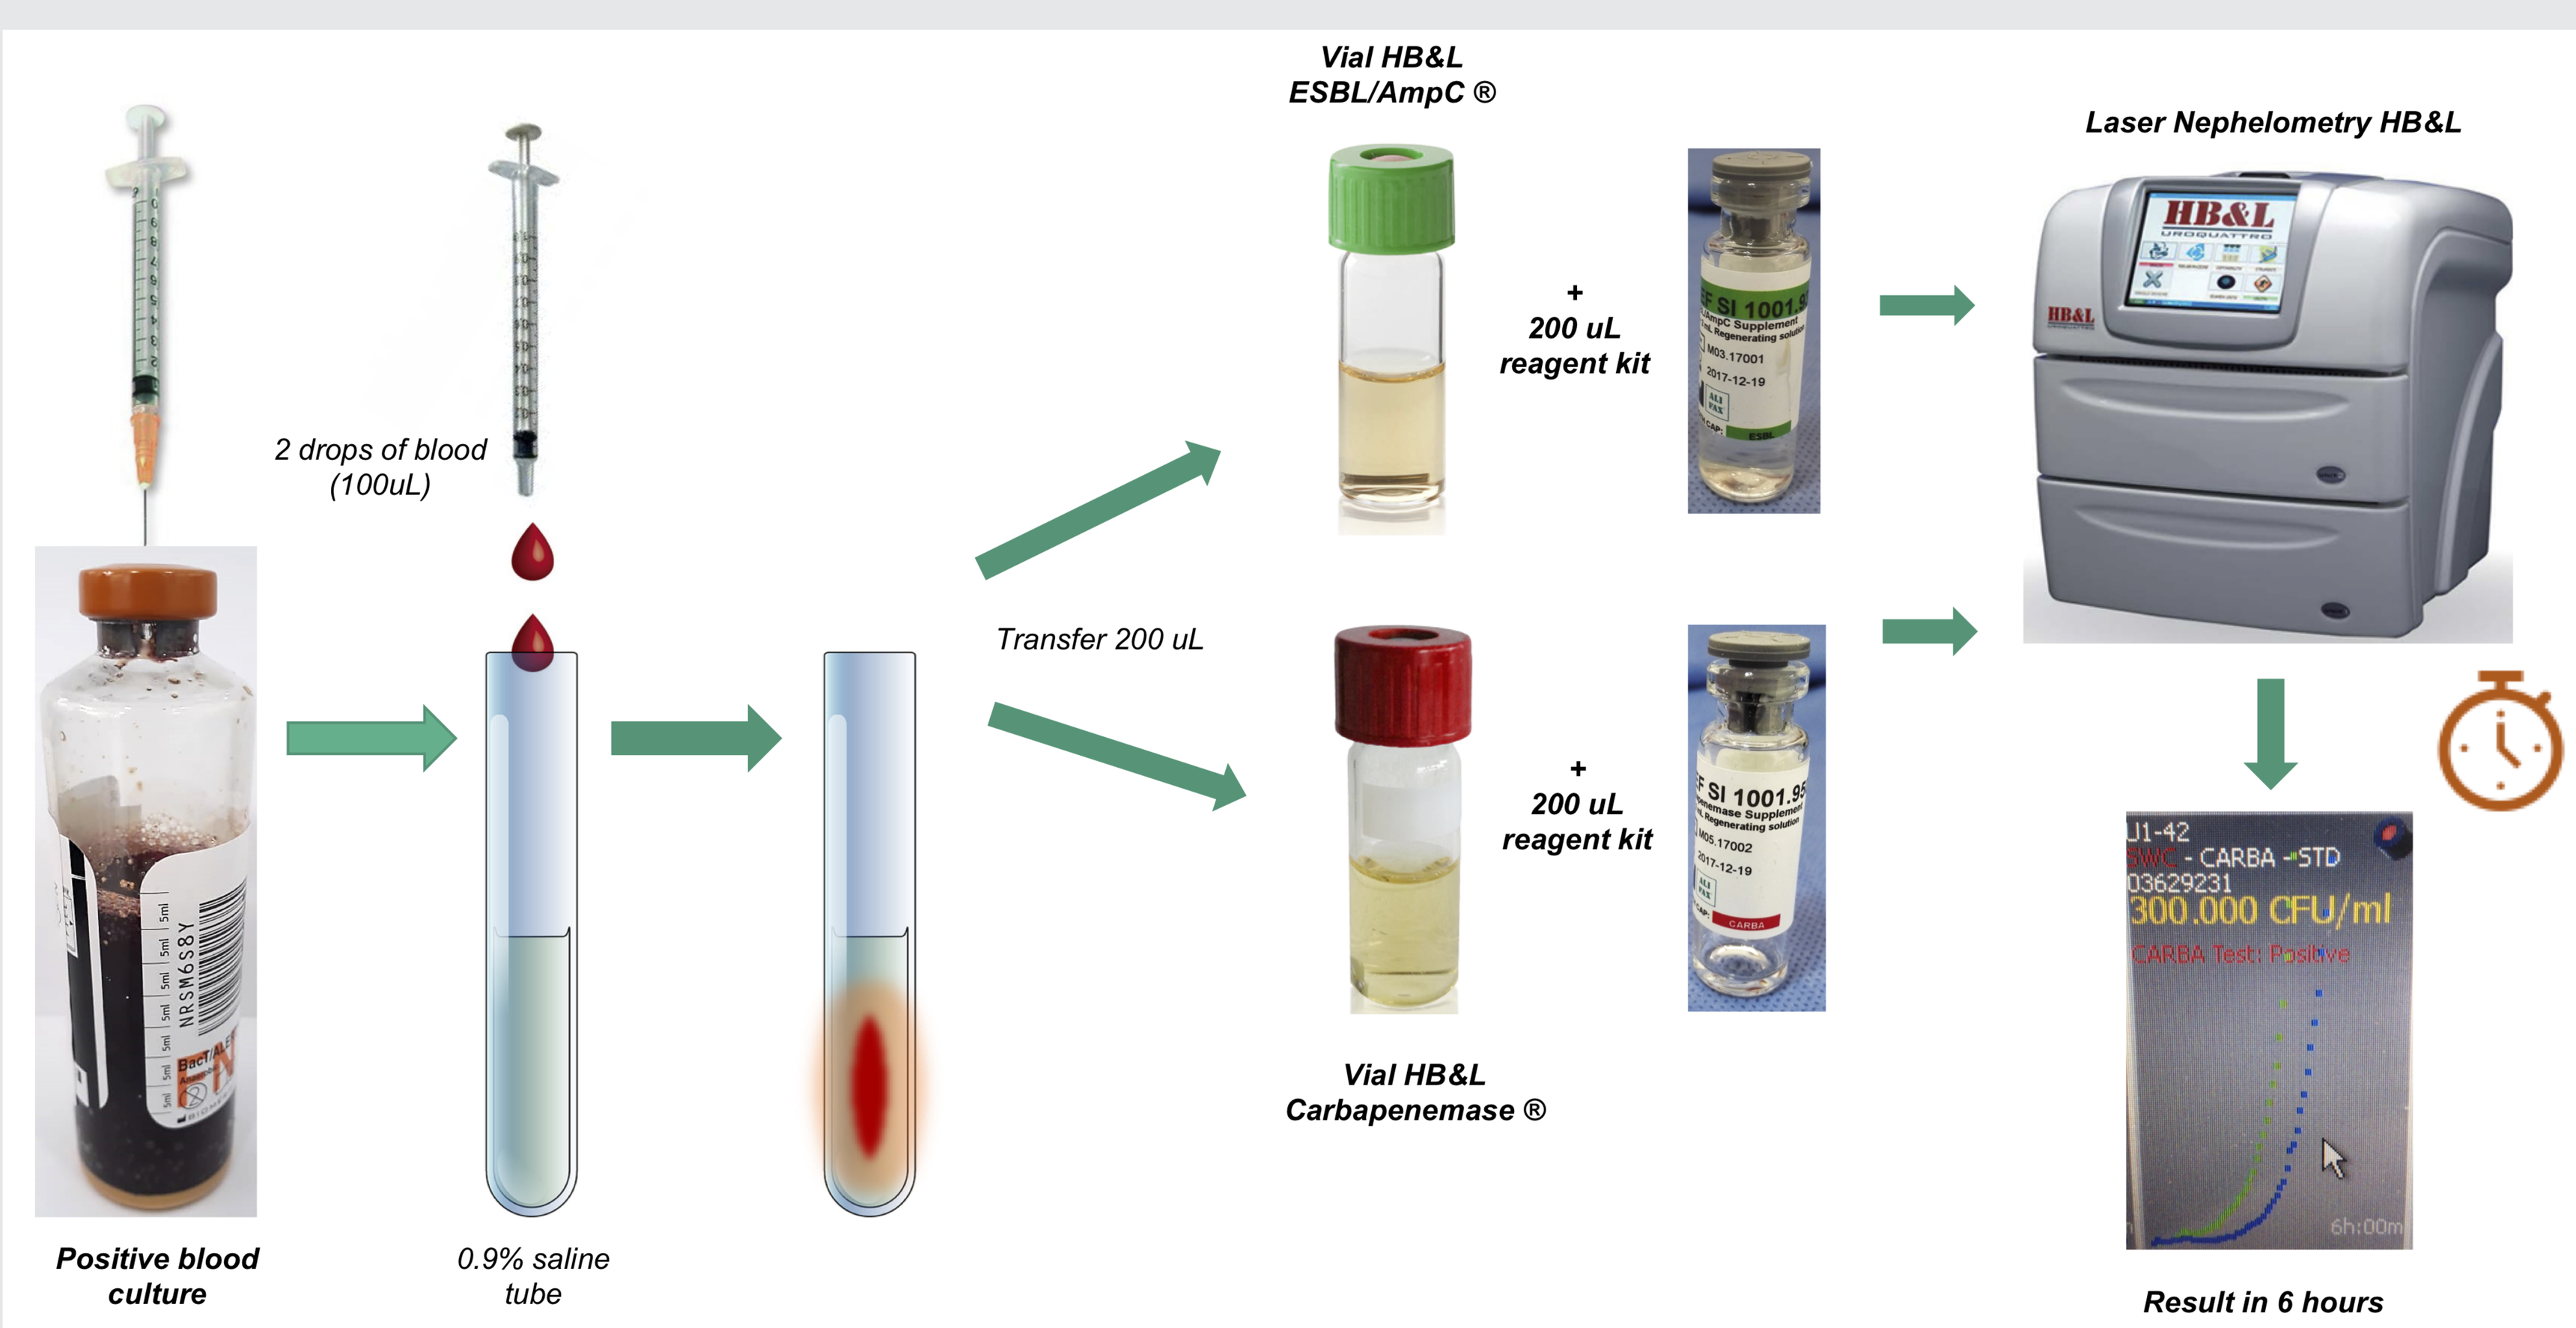
Vial HB&L
ESBL/AmpC ®
Laser Nephelometry HB&L
2 drops of blood (100uL)
Transfer 200 uL
+ 200 uL reagent kit
+ 200 uL reagent kit
Vial HB&L
Carbapenemase ®
300.000 CFU/ml
Positive blood culture
0.9% saline tube
Result in 6 hours

RESULTS

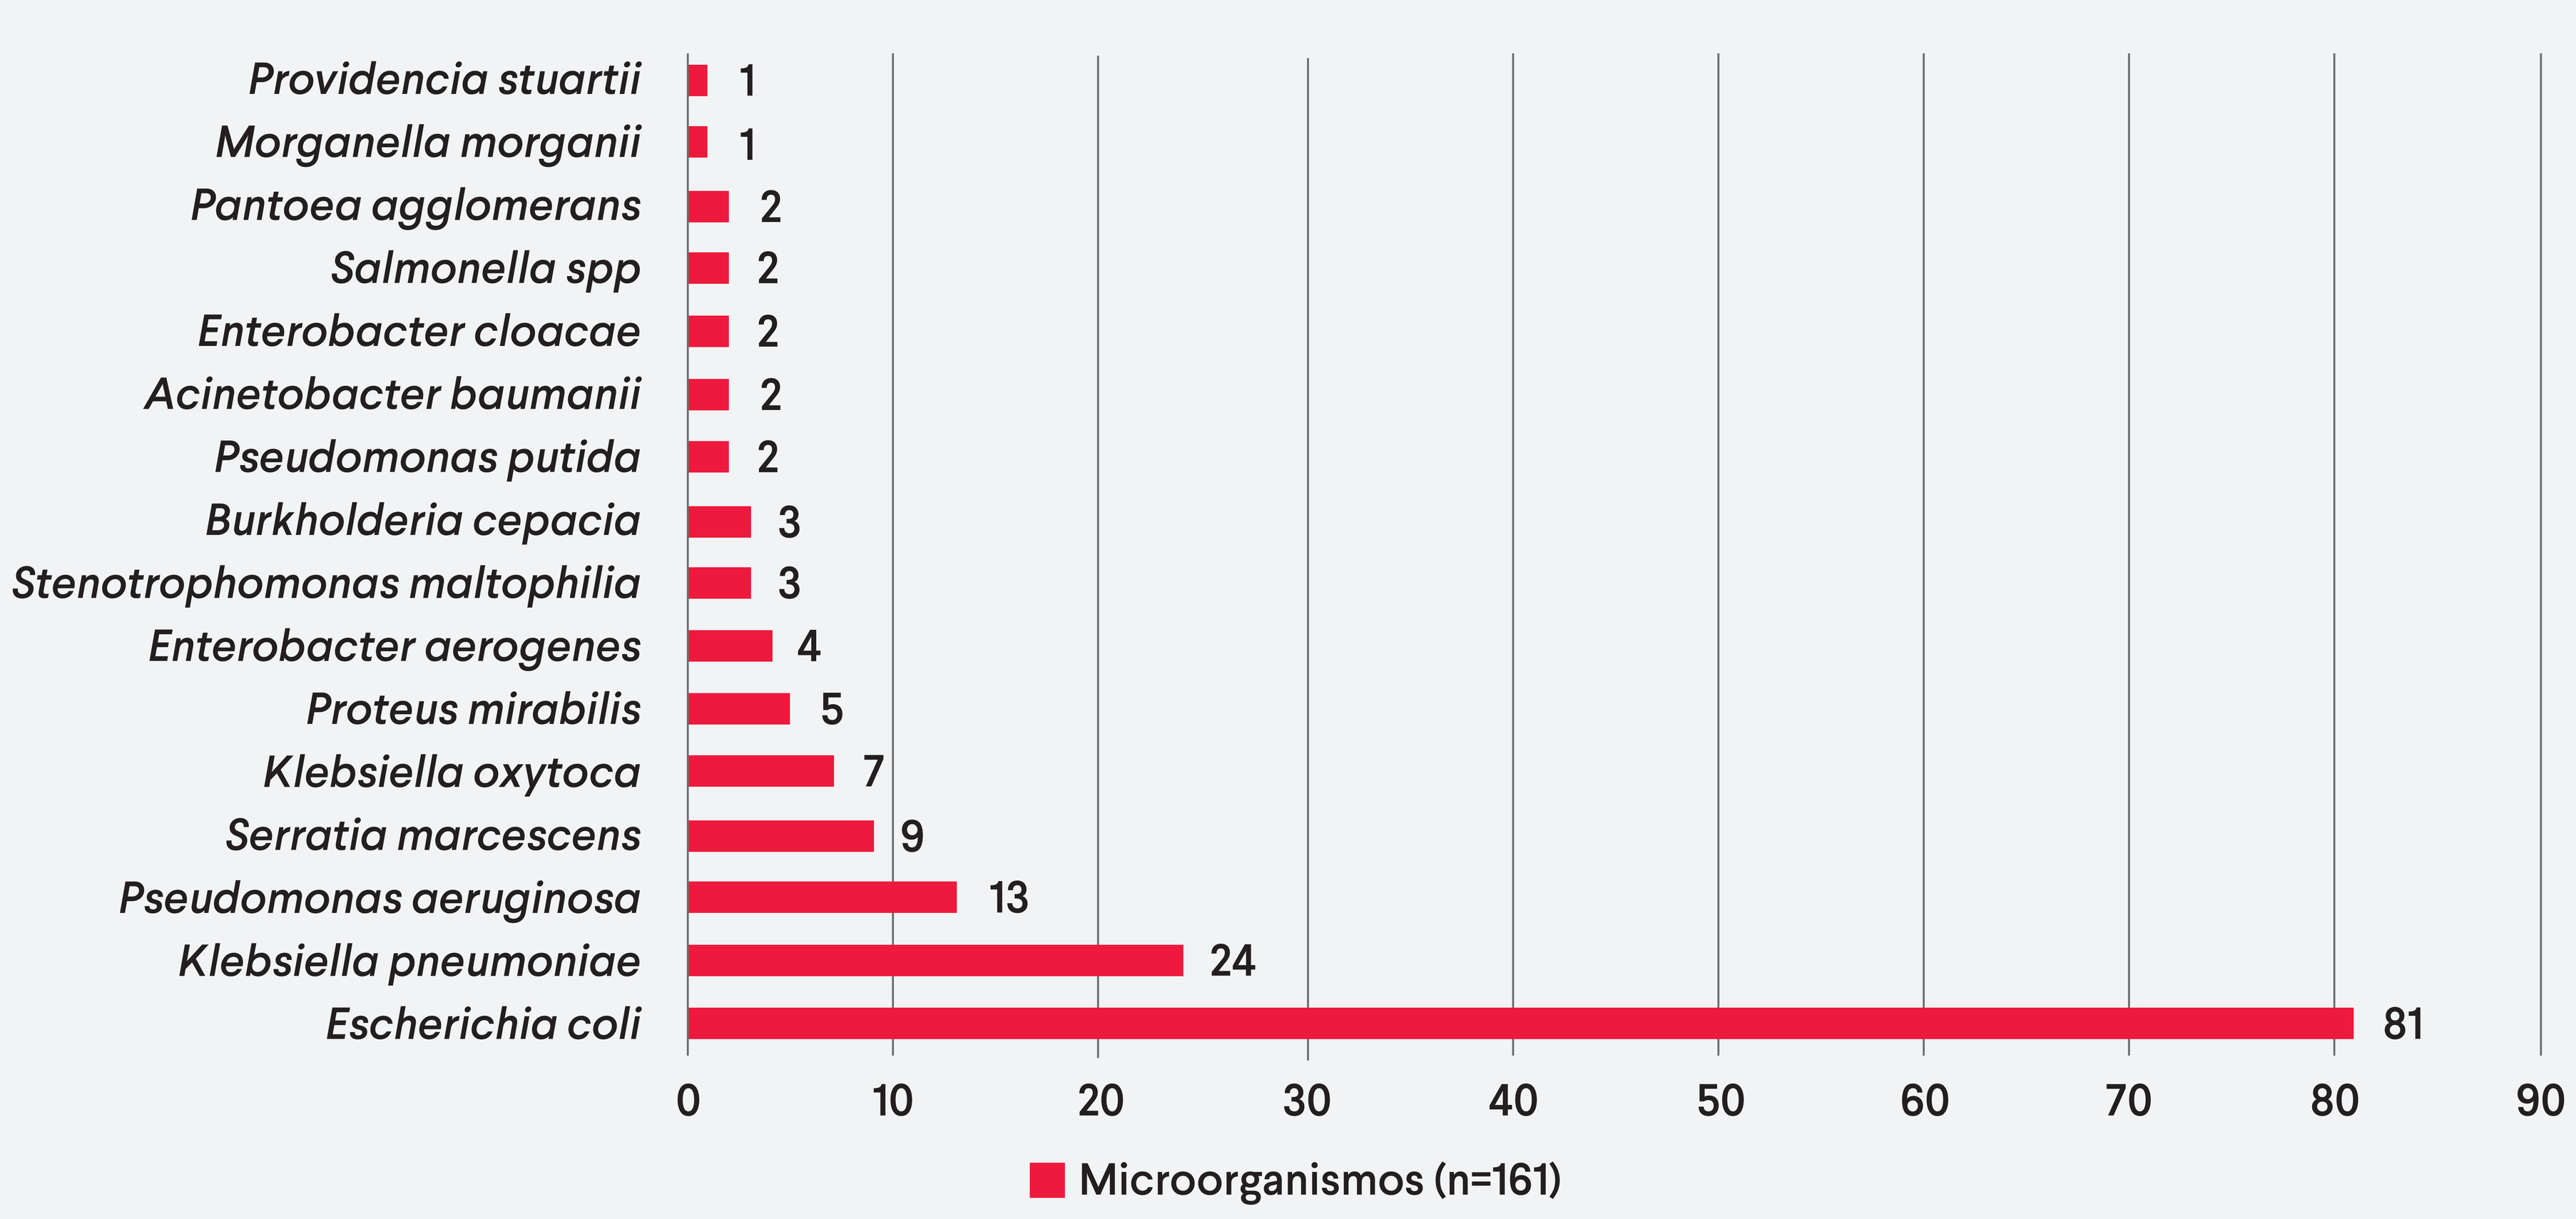
Providencia stuartii 1
Morganella morganii 1
Pantoea agglomerans 2
Salmonella spp 2
Enterobacter cloacae 2
Acinetobacter baumanii 2
Pseudomonas putida 2
Burkholderia cepacia 3
Stenotrophomonas maltophilia 3
Enterobacter aerogenes 4
Proteus mirabilis 5
Klebsiella oxytoca 7
Serratia marcescens 9
Pseudomonas aeruginosa 13
Klebsiella pneumoniae 24
Escherichia coli 81
0 10 20 30 40 50 60 70 80 90
Microorganismos (n=161)

	Vial ESBL/AmpC®				Vial Carbapenemase®			
	Positive	Negative	Total		Positive	Negative	Total	
Positive conventional culture	61	2	63	S (95%)	34	0	34	S (100%)
Negative conventional culture	0	98	98	E (100%)	0	127	0	E (100%)
Total	61	100	161		34	127	161	
	PPV 100%	NPV 98%			PPV 100%	NPV 100%		

S: sensitivity; E: specificity; PPV: Positive predictive value; VPN: Negative Predictive Value

RESULTS

- Concordance with traditional method was 99% (159/161)
- Identification times were significantly shorter (Median [IQR]; 19 hours [18, 22] Vs 61 hours [60, 64], p<0.001)
- 33% (53/161) isolations usual resistance patters
- 21% (34/161) were producer of carbapenemases and 13% (21/161) of extended-spectrum ß-lactamases.

	Microorganisms	(n)	Conventional Method / HB&L Method						
			Sensitive	IRT	BLEA	ESBL	AmpC repressed	AmpC repressed	Carbapenem resistant
Enterobacteria	*E. Coli*	81	34/34	13/13	15/15	17/17			2/2
	K. pneumoniae	24	6/6		1/1	2/2			15/15
	S. marcescens	9					4/4	5/5	
	K. oxytoca	7	4/4		2/2	1/1			
	P. mirabilis	5	5/5						
	E. aerogenes	4					1/1	1/1	2/2
	E. cloacae	2						2/2	
	Salmonella spp	2	1/1			1/1			
	P. aglomerans	2							2/2
NonFermenters	*M. morganii*	1						1/1	
	P. stuartii	1					1/1		
	P. putida	2							2/2
	P. aeruginosa	13					5/5	3/1**	5/5
	B. cepacea	3	2/2						1/1
	S. maltophilia	3							3/3
	A. baumanii	2							2/2

CONCLUSIONS

- Using our HB&L® modified protocol is an effective strategy to reduce the time to MDRP detection.
- Faster results, at a lower cost and with high concordance

REFERENCES

1. Tumbarello M, et al., 2007. Predictors of mortality in patients with bloodstream infections caused by extended-spectrum-beta-lactamase-producing Enterobacteriaceae: importance of inadequate initial antimicrobial treatment. Antimicrob Agents Chemother. 2007;51(6).

2. Giske CG et al., 2008. Clinical and economic impact of common multidrug-resistant Gram-negative bacilli. Antimicrob Agents Chemother. 2008;52(3):813-21